MICROCRYSTALLINE TYROSINE (MCT), A PROMISING DEPOT-FORMING ADJUVANT FOR EFFECTIVE T CELL RESPONSE IN VLP-BASED VACCINES IN MELANOMA MURINE MODELS

Mohsen M.O.[1,2], Lipp C.[2], Heath M.D.[3], Skinner M.A.[3], Kramer M.F.[3], Stein J.V.[4], Bachmann M.[1,2]

[1]Jenner Institute, Nuffield Department of Medicine, University of Oxford, United Kingdom [2]Inselspital, Universitatslinik RIA, Immunologie, Bern, Switzerland

[3]Allergy Therapeutics, Worthing, United Kingdom [4]Theodor Kocher Institute, University of Bern, Bern, Switzerland

Allergy Therapeutics® Transforming Allergy Treatment

UNIVERSITÄT BERN

UNIVERSITY OF OXFORD

INTRODUCTION

A rational design for an effective cancer vaccine may consider data derived the following key components: cancer antigens (Ag), and adjuvants. Depot adjuvants enhance vaccine efficacy mainly by prolonging Ag presentation for a duration optimal for T cell clonal expansions. Aluminium (Alum) is one of the limited adjuvants approved for use in humans, however Alum may be suboptimal for use in anti-tumor activity when combined with cancer vaccines as it shifts the immune response towards a more biased Th2 profile. Microcrystalline Tyrosine (MCT) is a proprietary depot adjuvant which is used in allergen-specific immunotherapy. MCT has several advantages over Alum including being a degradable adjuvant which will efficiently be cleared from the body. The current project aims to study the efficacy of MCT in combination with virus-like particle (VLP) based vaccines in inducing T cell response in melanoma murine models and to compare its efficacy to the widely used Alum adjuvant.

METHOD

Schematic diagram illustrating tumor transplantation methodology. C57Bl/6 RAG-/- mice were used to establish B16F10p33 tumors. 14 days later the tumors were dissected and processed to 2mm pieces for transplantation in C57Bl/6 mice.

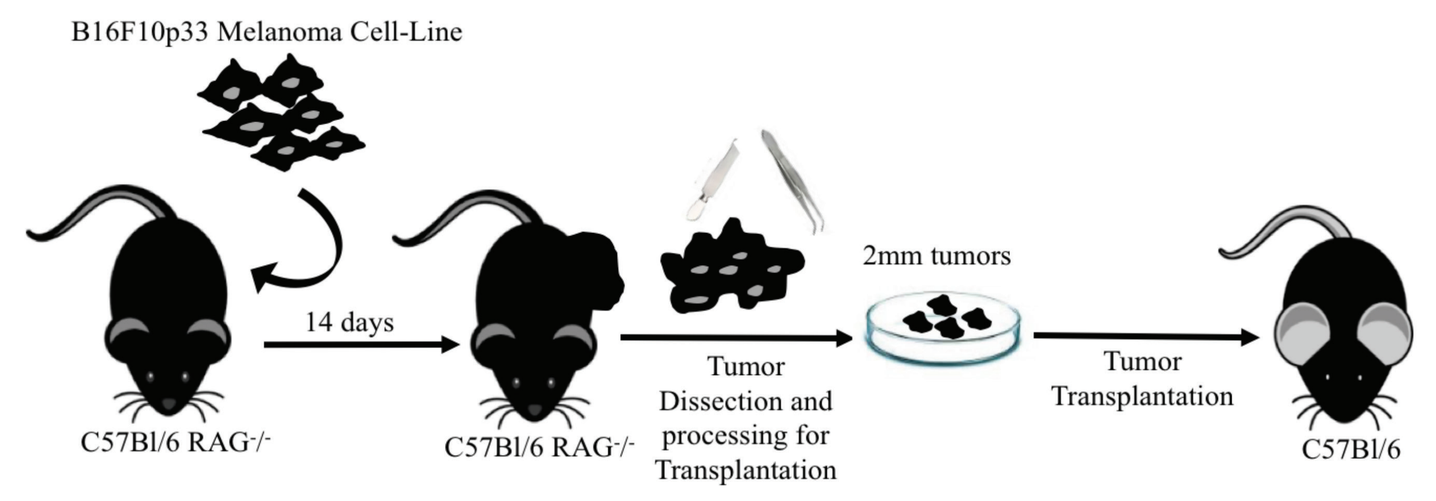

B16F10p33 Melanoma Cell-Line
14 days
C57Bl/6 RAG-/-
C57Bl/6 RAG-/-
Tumor Dissection and processing for Transplantation
2mm tumors
Tumor Transplantation
C57Bl/6

Vaccination regimen, CuMV-p33 (Cucumber Mosaic Virus) vaccine was tested separately or formulated in different adjuvants (Type-B CpGs – MCT or Alum) and injected s.c. at d4, d8 and d12 4 days post tumor transplantation.

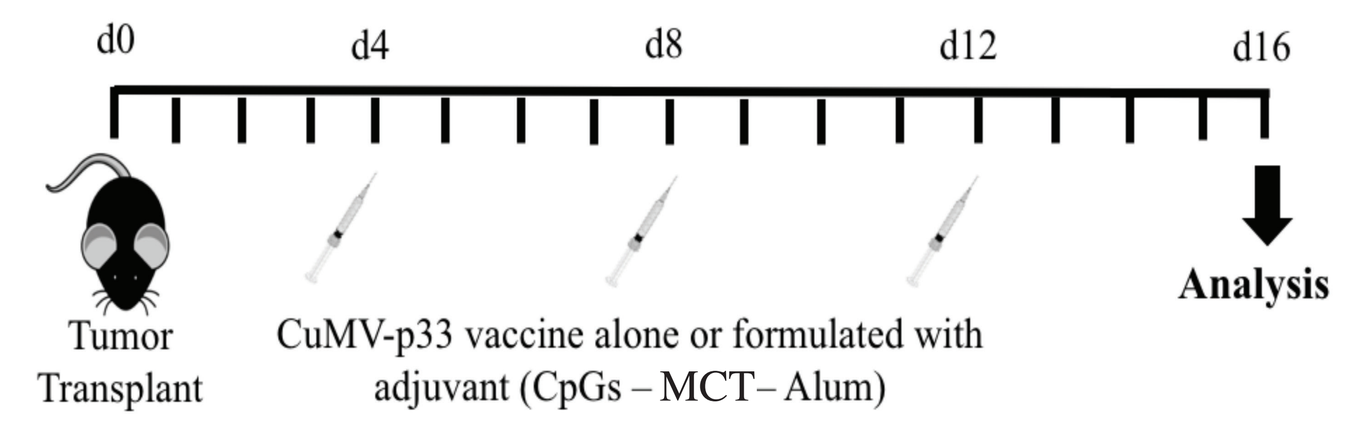

d0
d4
d8
d12
d16
Tumor Transplant
CuMV-p33 vaccine alone or formulated with adjuvant (CpGs – MCT– Alum)
Analysis

RESULTS

CuMV-VLPs an attractive cancer vaccine platform

- **Cu-MV-VLPs can efficiently drain into lymph nodes (LN)**

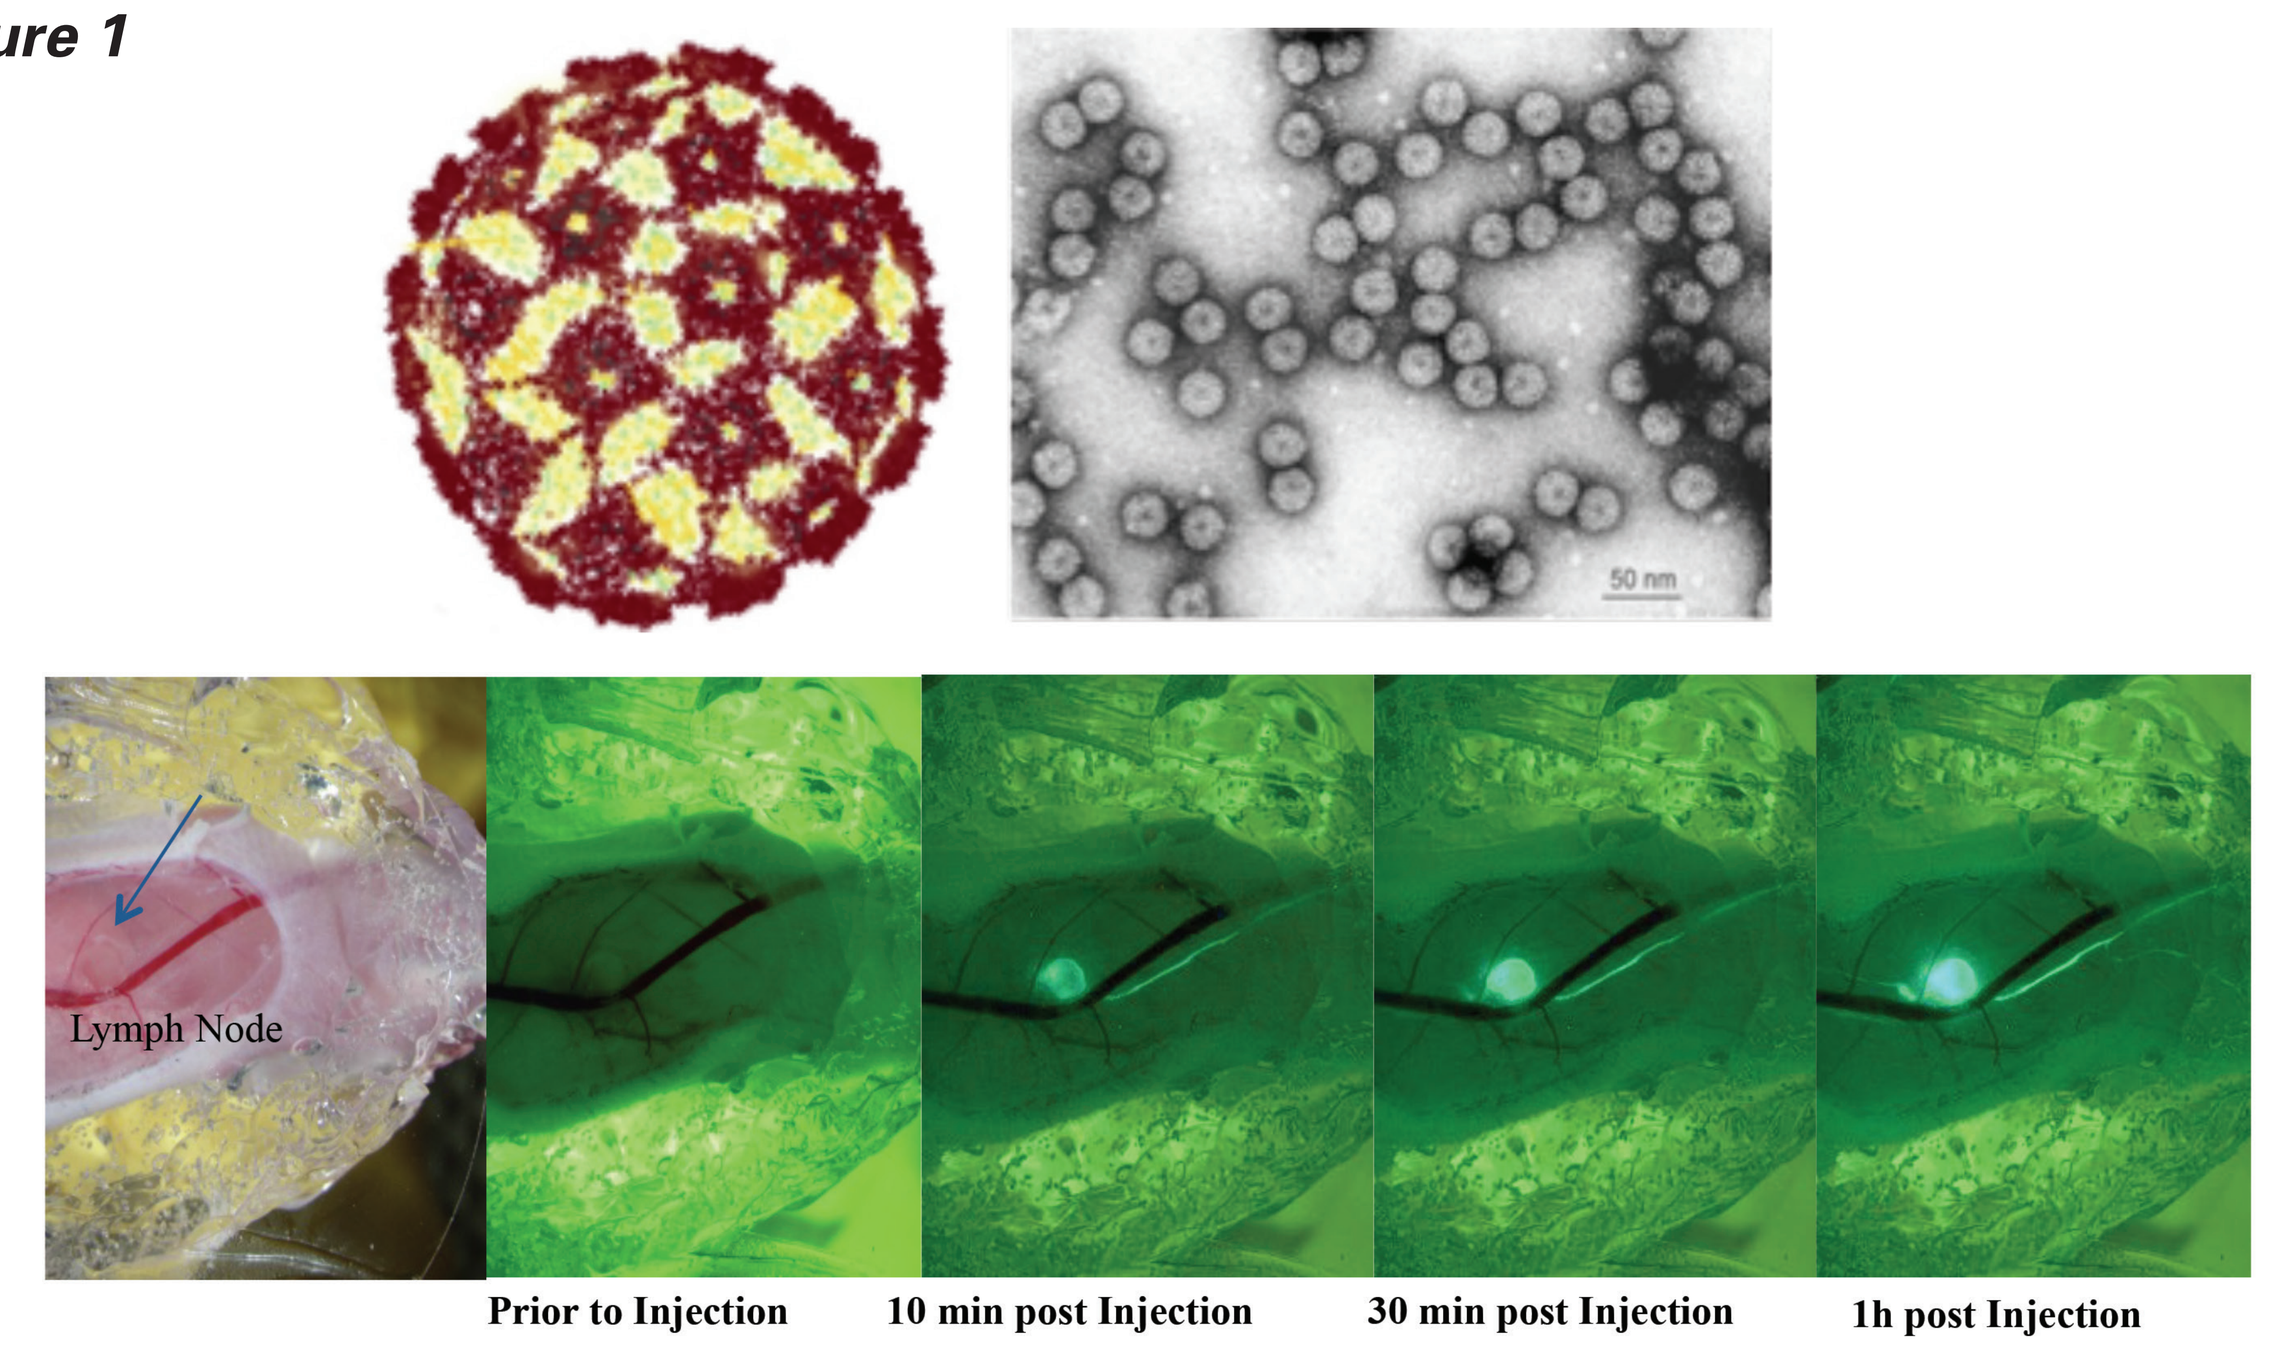

Figure 1
A
50 nm
B
Lymph Node
Prior to Injection
10 min post Injection
30 min post Injection
1h post Injection

Figure 1 **A,** CuMV-VLPs sketch and electron microscopy. **B,** Steriomicroscopic imaging illustrate the rapid drainage of CuMV-VLPs to the popliteal LN (less than 10min) when injected in mouse footpad

- **p33 derived from LCMV can efficiently be coupled to CuMV-VLPs by copper-free click chemistry**

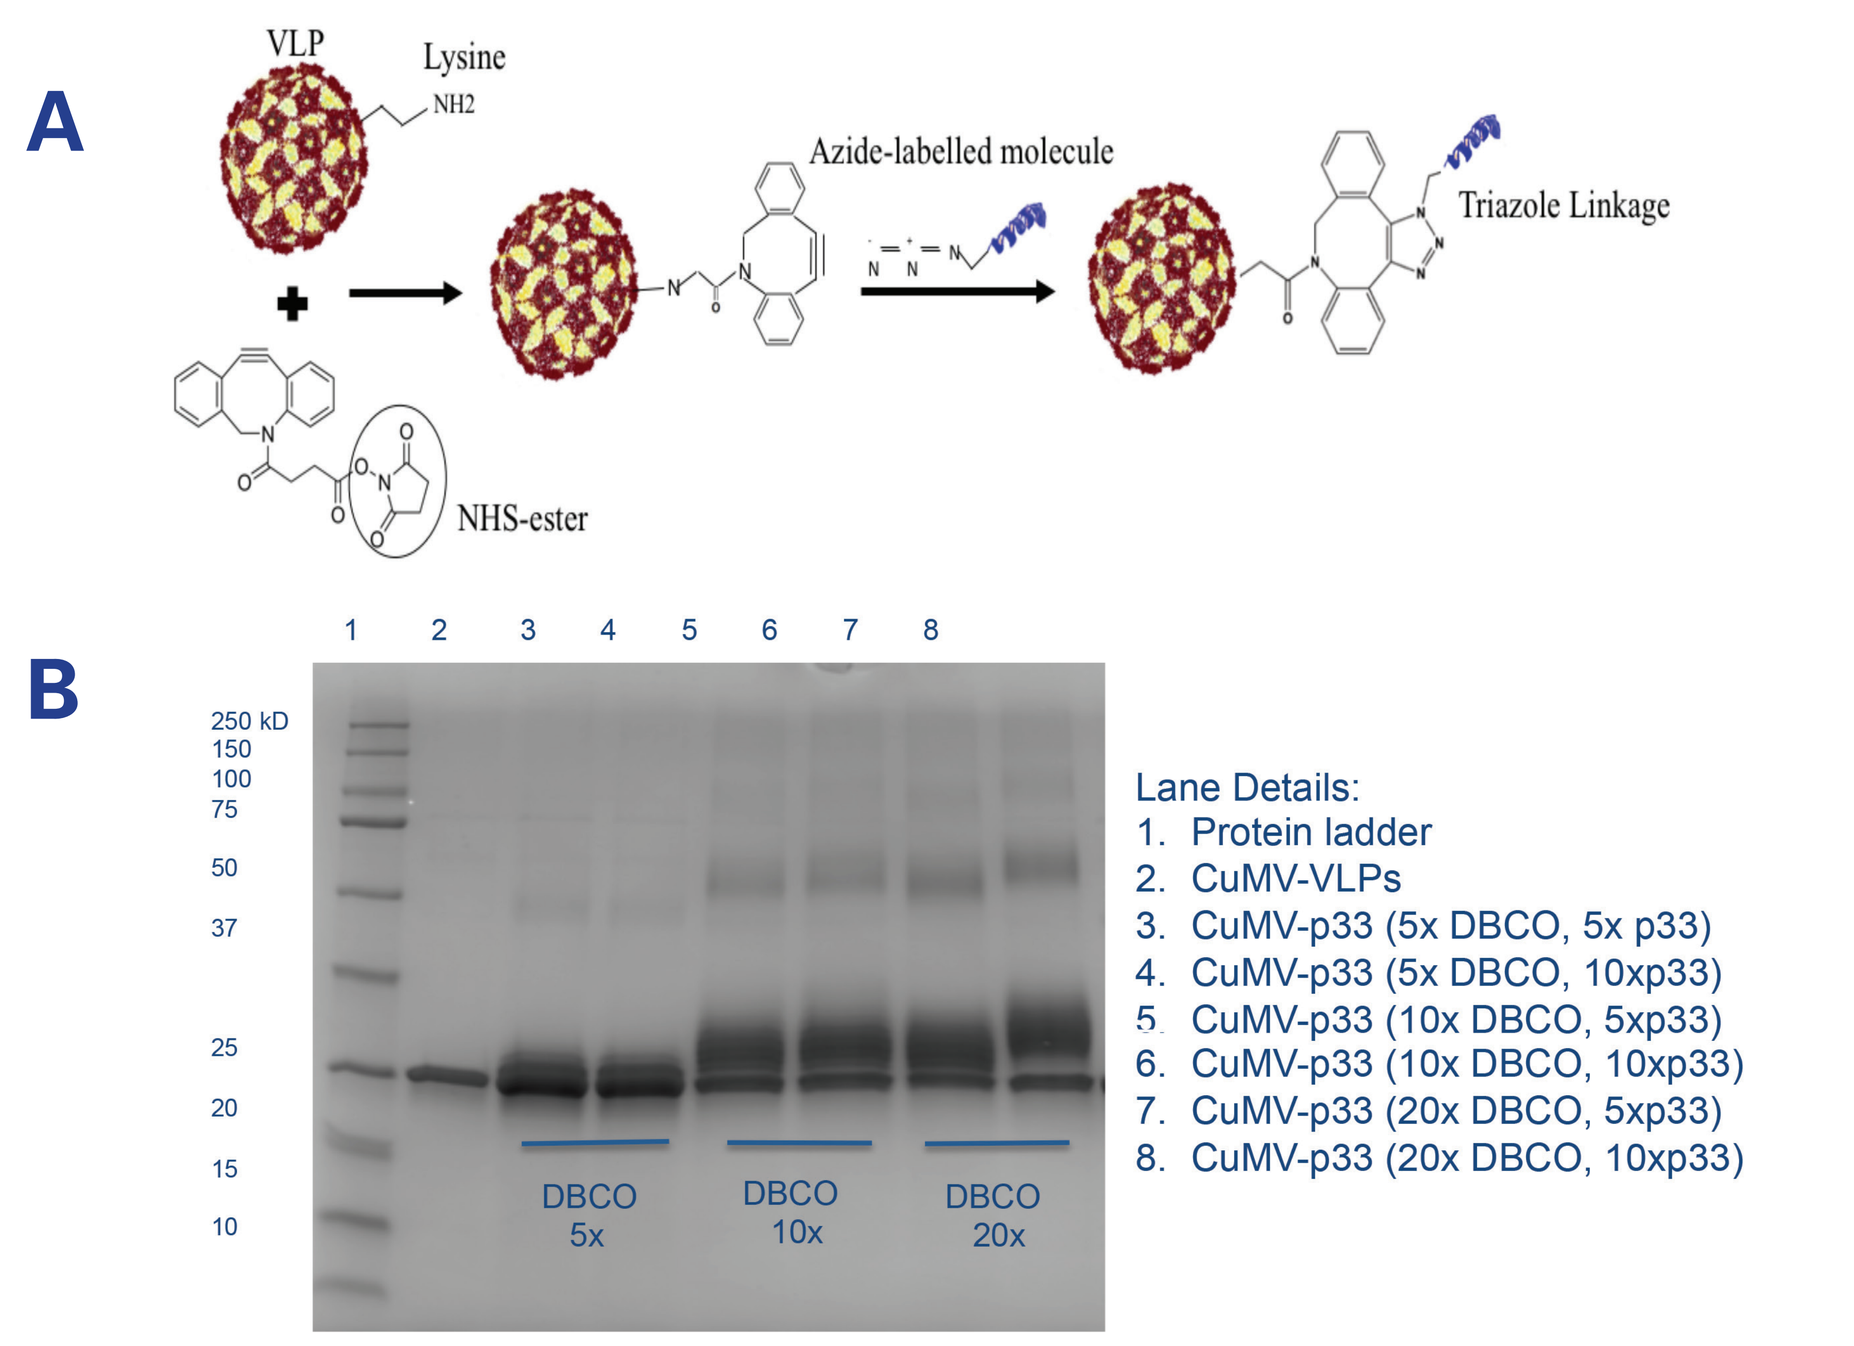

Figure 2
A
VLP
Lysine
NH2
Azide-labelled molecule
Triazole Linkage
NHS-ester
B
1 2 3 4 5 6 7 8
250 kD
150
100
75
50
37
25
20
15
10
DBCO 5x
DBCO 10x
DBCO 20x
Lane Details:
1. Protein ladder
2. CuMV-VLPs
3. CuMV-p33 (5x DBCO, 5x p33)
4. CuMV-p33 (5x DBCO, 10xp33)
5. CuMV-p33 (10x DBCO, 5xp33)
6. CuMV-p33 (10x DBCO, 10xp33)
7. CuMV-p33 (20x DBCO, 5xp33)
8. CuMV-p33 (20x DBCO, 10xp33)

Figure 2 **A,** schematic diagram illustrating the coupling method using cupper-free click chemistry. **B,** SDS-PAGE stained with Comassie Blue showing the coupling efficacy of p33 model antigen to CuMV-VLPs.

MCT shows depot effect when combined with CuMV-p33 vaccine

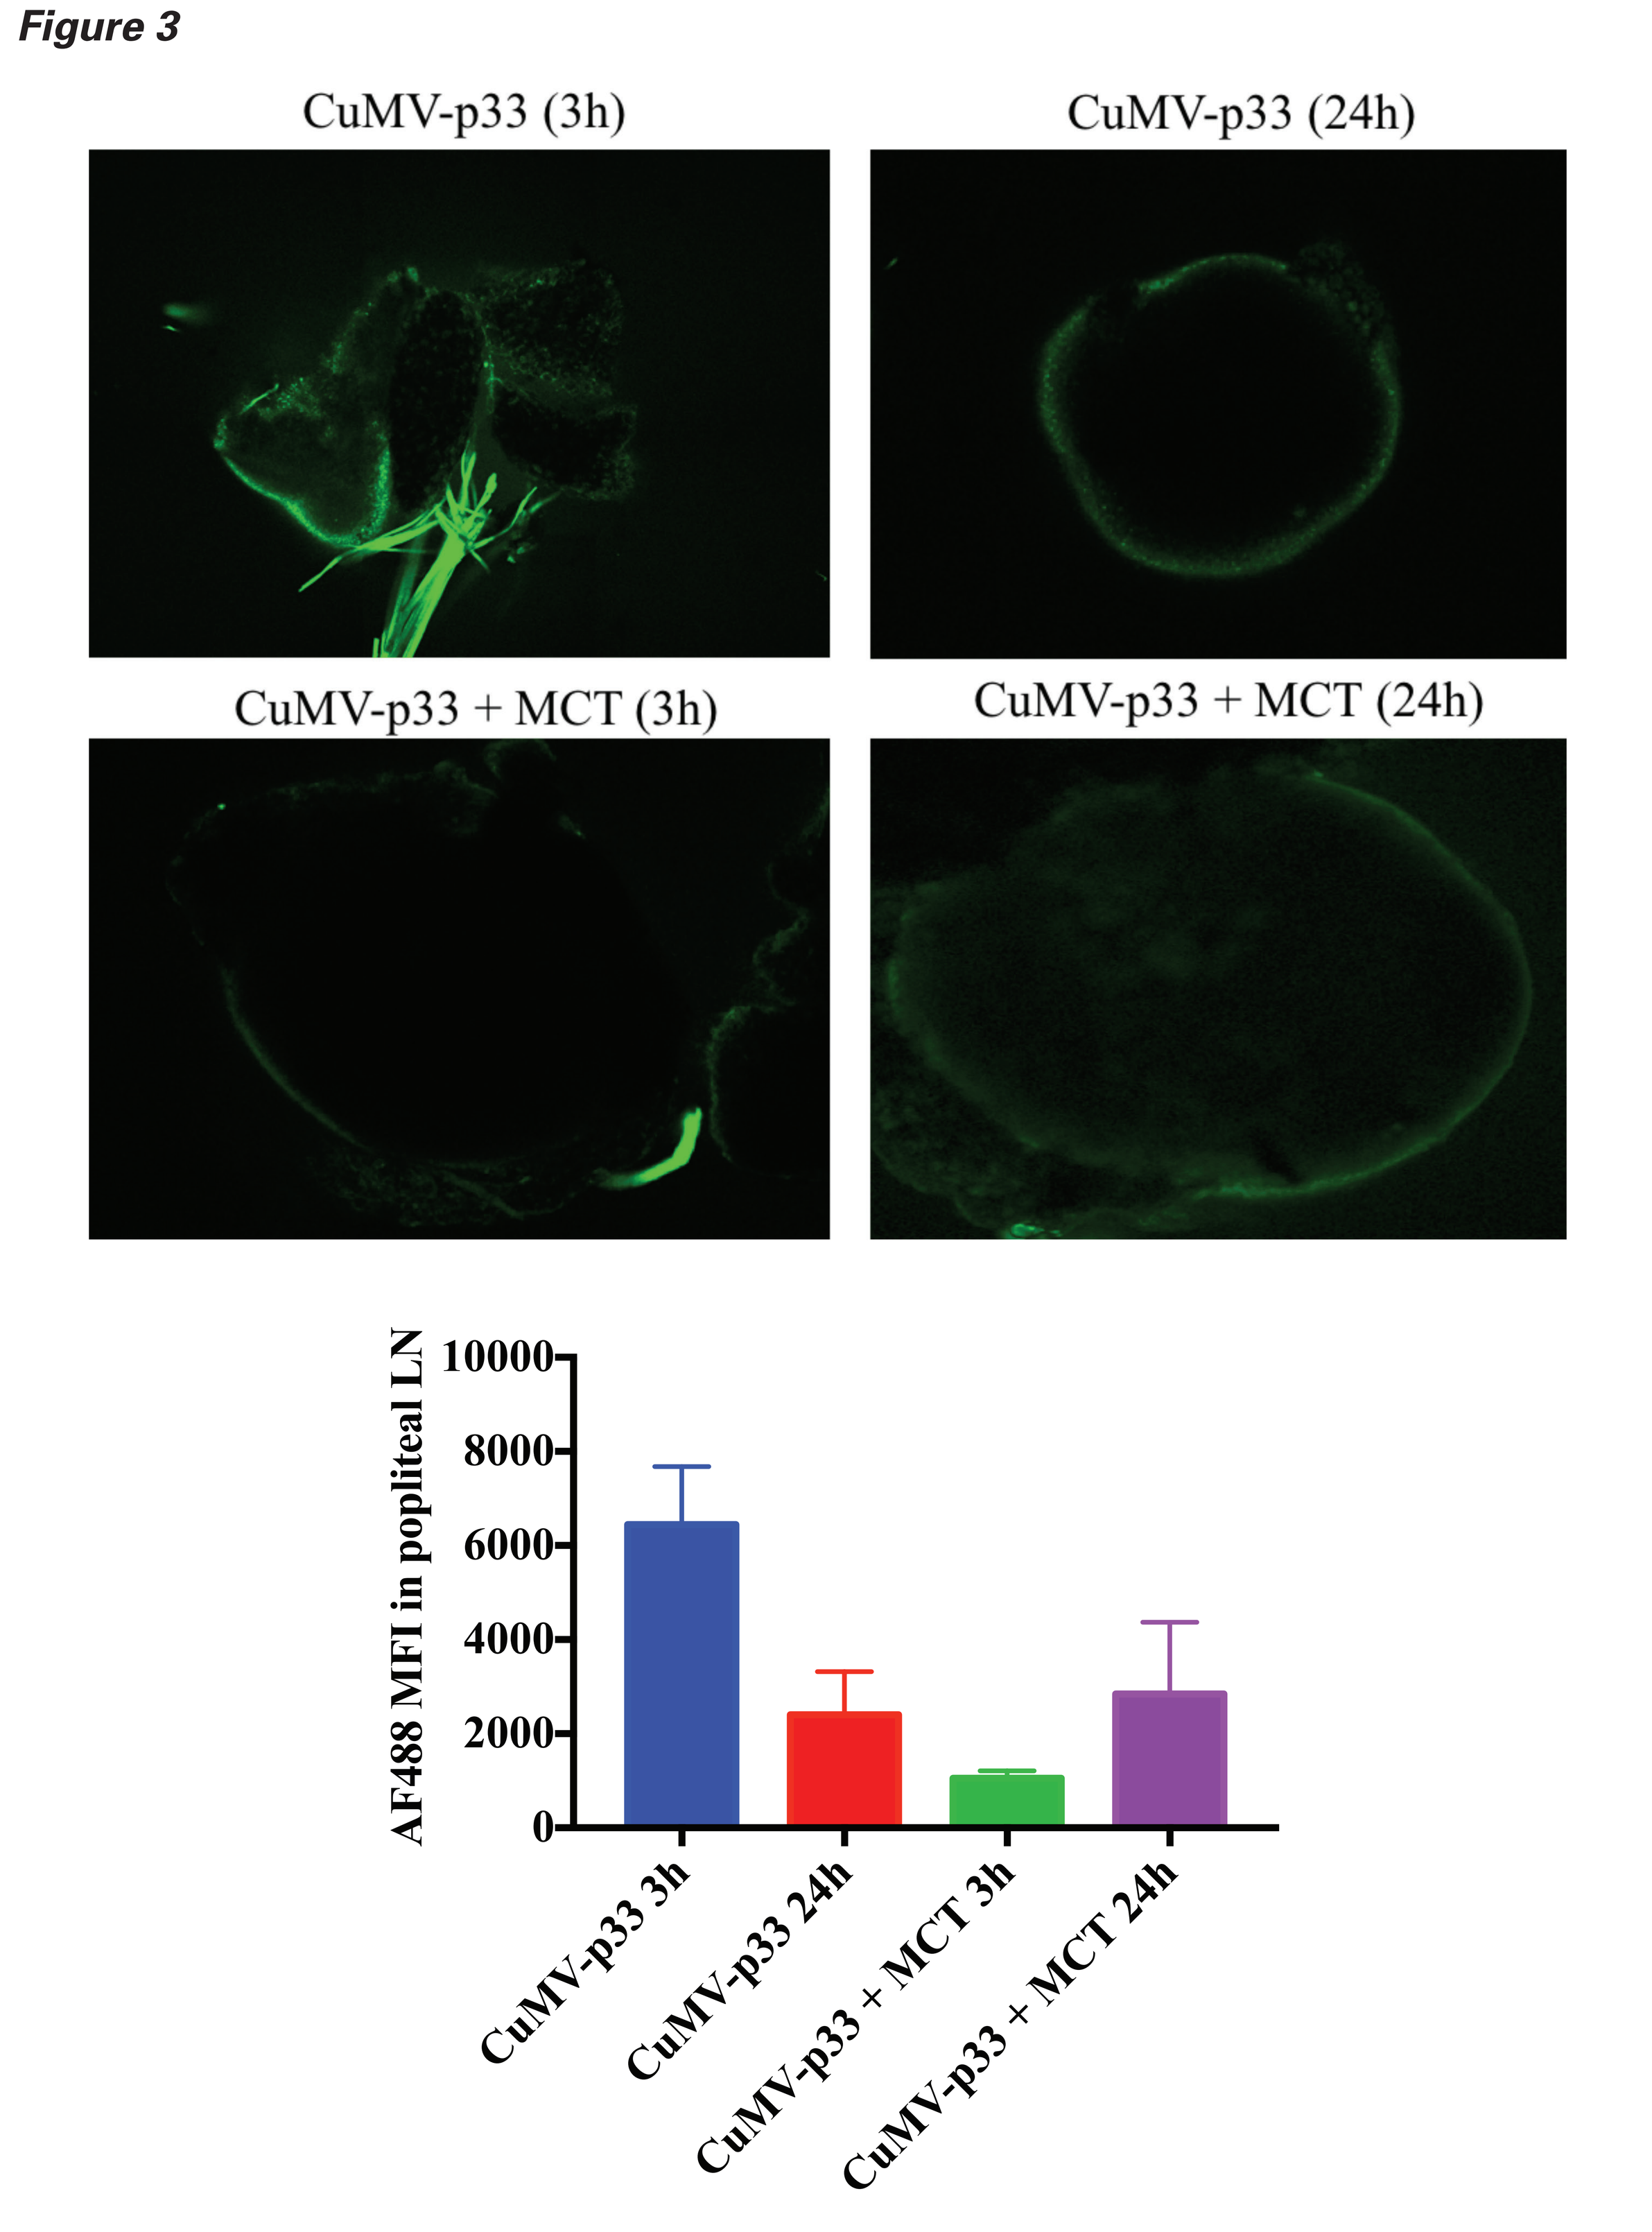

Figure 3
CuMV-p33 (3h)
CuMV-p33 (24h)
CuMV-p33 + MCT (3h)
CuMV-p33 + MCT (24h)
AF488 MFI in popliteal LN
10000
8000
6000
4000
2000
0
CuMV-p33 3h
CuMV-p33 24h
CuMV-p33 + MCT 3h
CuMV-p33 + MCT 24h

Figure 3 Confocal microscopy imaging of mouse popliteal LN 3h or 24h post injection of CuMV-p33 vaccine labelled with AF488 without or with MCT adjuvant s.c. in C57Bl/6 mice. Note the slow and prolonged release of CuMV-p33 vaccine when formulated with MCT.

MCT causes tumor regression and significantly increases p33 specific T cells, IFN-γ and TNF-α when formulated with CuMV-p33 vaccine in B16F10 melanoma murine models and shows superiority over Alum.

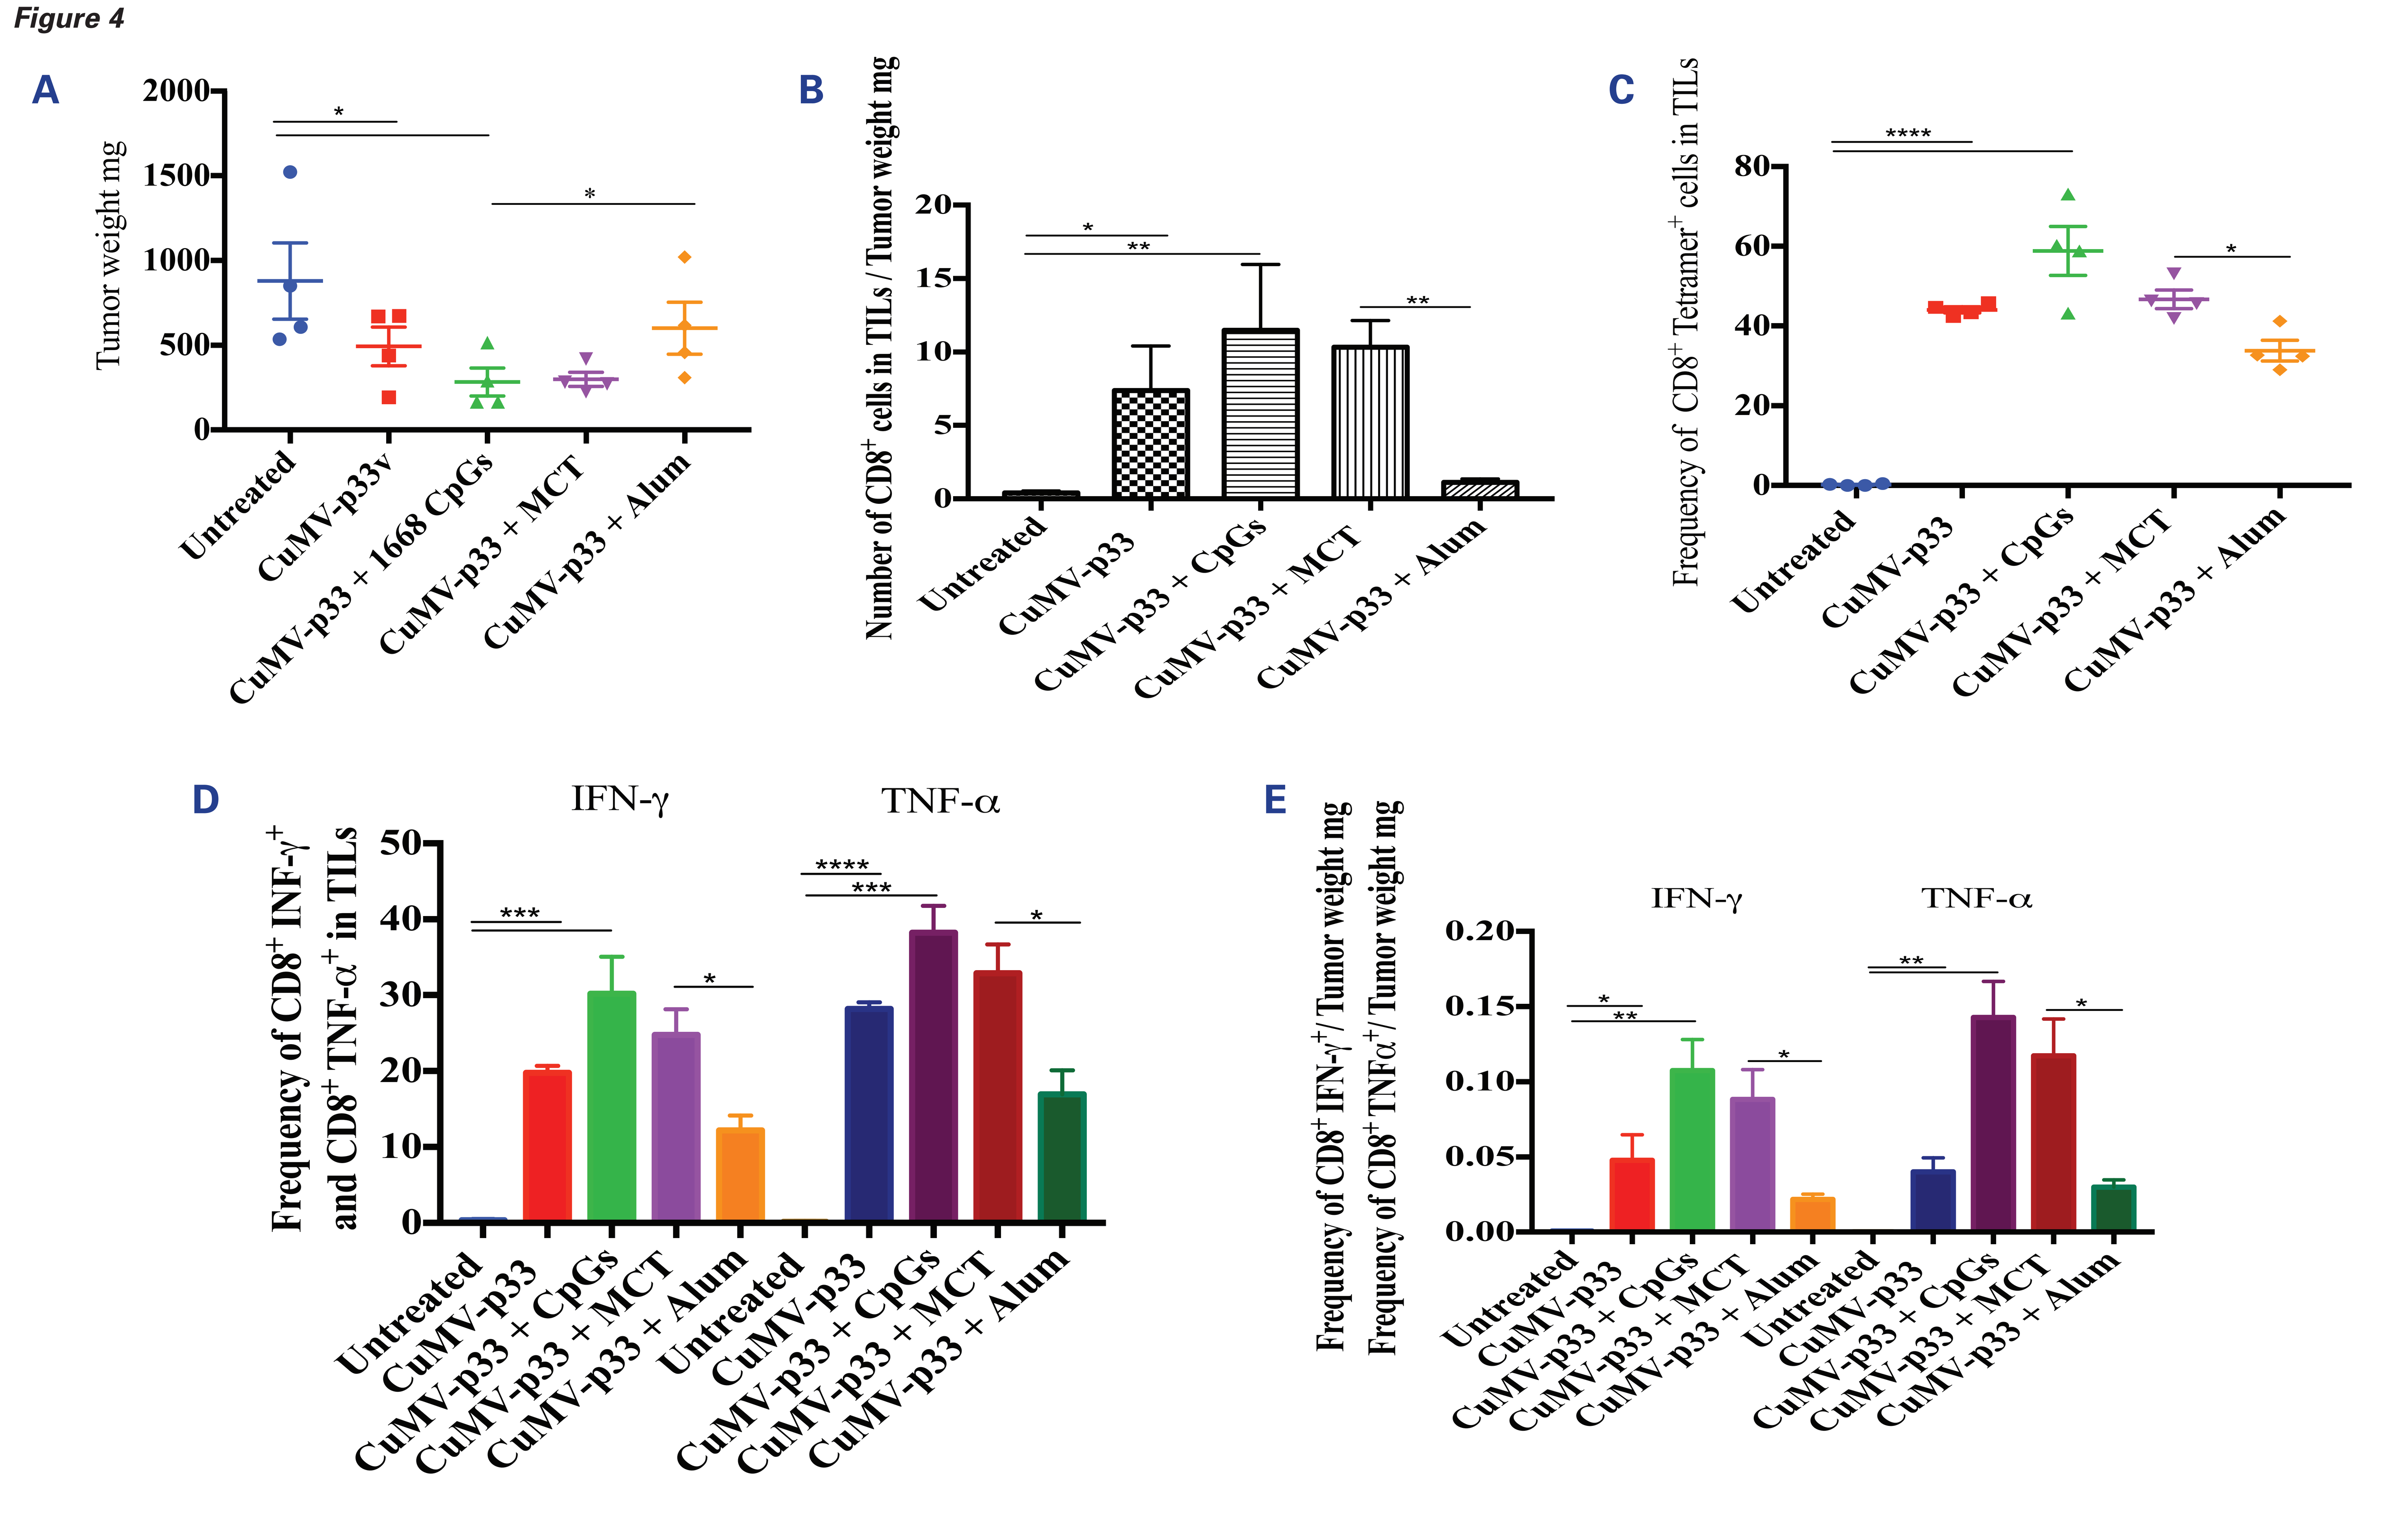

Figure 4
A
Tumor weight mg
2000
1500
1000
500
0
Untreated
CuMV-p33v
CuMV-p33 + 1668 CpGs
CuMV-p33 + MCT
CuMV-p33 + Alum
B
Number of CD8+ cells in TILs / Tumor weight mg
20
15
10
5
0
Untreated
CuMV-p33
CuMV-p33 + CpGs
CuMV-p33 + MCT
CuMV-p33 + Alum
C
Frequency of CD8+ Tetramer+ cells in TILs
80
60
40
20
0
D
IFN-γ
TNF-α
Frequency of CD8+ INF-γ+ and CD8+ TNF-α+ in TILs
50
40
30
20
10
0
E
IFN-γ
TNF-α
Frequency of CD8+ IFN-γ+/ Tumor weight mg
Frequency of CD8+ TNFα+/ Tumor weight mg
0.20
0.15
0.10
0.05
0.00

Figure 4 **A,** Tumor weight (mg) in each designated group. **B,** Ratio of CD8⁺ cells in TILs/Tumor weight in each designated group. **C,** Frequency of CD8⁺ Tetramer⁺ cells in TILs in each designated group. (4 mice per group), one representative of 3 similar experiments is shown. **D,** Frequency of CD8⁺ IFN-γ and CD8⁺ TNF-α⁺ cells in TILs in each designated group. **E,** Ratio of CD8⁺ IFN-γ and CD8⁺ TNF-α⁺ in TILs/ Tumor weight in each designated group. (4 mice per group), one representative of 3 similar experiments is shown.

REFERENCES

- Ammi, R., *et al.* Poly(I:C) as cancer vaccine adjuvant: Knocking on the door of medical breakthroughs. Pharmacology & Therapeutics (2015),146, 120-131.
- Bell, A. J., *et al.* The adsorption of allergoids and 3-O-desacyl-4 '-monophosphoryl lipid A (MPL) to microcrystalline tyrosine (MCT) in formulations for use in allergy immunotherapy. Journal of Inorganic Biochemistry (2015), 152, 147-153.
- Klimek L., *et al.* Clinical use of adjuvants in allergen-immunotherapy. Expert Review of clinical immunology, 2017. Vol. 13, No. 6, 599-610.

CONCLUSIONS

MCT is a promising depot-forming adjuvant which enhances cytotoxic T cell (CTL) response in murine models. Formulating a CuMV-VLPs vaccine with MCT has shown significant regression in tumor weight comparable to Type-B CpGs adjuvants. Furthermore, p33 specific T cell response as well as IFN-γ and TNF-α production was significantly enhanced when using MCT. On the other hand, combining CuMV-p33 vaccine with Alum did not cause tumor regression and did not result in significant CTL response. Hence, in contrast to Alum, MCT is an adjuvant enhancing CTL response in mice.

IM/GEN/E/2018/017

EAACI_2018_Poster_12_V2.indd 1 13/04/2018 08:36